Abstract 3102

Phase 1b/2 SEASTAR Trial: Safety, Pharmacokinetics, and Preliminary Efficacy of the Poly(ADP-ribose) Polymerase (PARP) Inhibitor Rucaparib and Angiogenesis Inhibitor Lucitanib in Patients With Advanced Solid Tumors

Ecaterina E. Dumbrava,[1] Geoffrey Shapiro,[2] Johanna Bendell,[3] Timothy A. Yap,[1] Rinath Jeselsohn,[2] Denise Lepley,[4] Sabrina Hurley,[4] Kevin K. Lin,[4] Mingxiang Liao,[4] Jenn Habeck,[4] Aaron Enke,[4] Erika Hamilton[3]

[1]The University of Texas MD Anderson Cancer Center, Houston, TX, USA; [2]Dana-Farber Cancer Institute, Boston, MA, USA; [3]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN, USA; [4]Clovis Oncology, Inc., Boulder, CO, USA

SUMMARY

- **Initial findings suggest that rucaparib + lucitanib has an acceptable safety profile**
- **Among patients with measurable disease, there was some evidence of effect on tumor and disease stabilization; the disease control rate was 23.5% (4/17)**
- **A maximum tolerated dose for the combination has not been established based on our data; lucitanib is currently being evaluated in phase 2 combination studies at a starting dosage of 6 mg once daily**
- **No drug-drug interactions were observed between lucitanib and rucaparib**
- **The combination of a poly(ADP-ribose) polymerase inhibitor and an angiogenesis inhibitor is feasible and may merit further evaluation**

Poster Download
Also available at: https://clovisoncology.com/files/ASCO2021_Dumbrava_Poster.pdf
Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without written permission from ASCO® and the author of this poster.
Corresponding author: *Ecaterina E. Dumbrava; EElleana@mdanderson.org.*

Poster Slides
Also available at: https://clovisoncology.com/files/ASCO2021_Dumbrava_Slides.pdf
Please scan this QR code or visit the website to view the poster as slides.

INTRODUCTION

- Rucaparib is an oral, potent, and selective inhibitor of poly(ADP-ribose) polymerase (PARP) 1, PARP2, and PARP3[1–3]
- Lucitanib is an oral tyrosine kinase inhibitor that selectively inhibits vascular endothelial growth factor receptors 1–3 (VEGFR1–3), platelet-derived growth factor receptors alpha and beta (PDGFR α/β), and fibroblast growth factor receptors 1–3 (FGFR1–3)[4]
- In preclinical studies, the antitumor activity of rucaparib was enhanced by lucitanib through antiproliferative, antiangiogenic, and immunological mechanisms[5]
- We hypothesize that combining lucitanib and rucaparib is tolerable and can induce a higher hypoxic state and homologous recombination repair deficiency[6–9] that may lead to greater sensitivity to PARP inhibition
- Here, we describe the safety, tolerability, pharmacokinetics (PK), and preliminary efficacy of rucaparib + lucitanib from Arm A of the phase 1b part of the SEASTAR study (NCT03992131), an open-label, parallel-arm study of rucaparib in combination with other anticancer agents

METHODS

- For phase 1b, patients with advanced solid tumors who had ≥1 prior line of therapy in the locally advanced, recurrent, or metastatic setting and who had progressed on standard therapy were eligible; patients with *BRCA1*- or *BRCA2*-mutated ovarian cancer must have received prior PARP inhibitor therapy
- A 3+3 design was used, with starting doses of rucaparib 300 mg twice daily and lucitanib 4 mg once daily in cohort 1
- Dose-limiting toxicities (DLTs) were assessed during the first 28 days of treatment
- Plasma samples were collected for PK analyses
- Genomic alterations were identified by local testing or through central testing of plasma or tumor tissue
- The primary endpoint of phase 1b was the incidence of grade 3 or 4 treatment-emergent adverse events (TEAEs) and clinical laboratory abnormalities defined as DLTs
 - A key secondary endpoint was investigator-assessed objective response rate per Response Evaluation Criteria In Solid Tumors, version 1.1 (RECIST)
 - A post hoc exploratory endpoint was the disease control rate, defined as the proportion of patients with confirmed investigator-assessed response (complete or partial response), or stable disease per RECIST lasting ≥16 weeks

RESULTS

- As of April 16, 2021, 19 patients were treated with rucaparib + lucitanib and included in the analyses (**Table 1**)

Table 1. Patient Demographics and Baseline Characteristics

	Cohort 1: Rucaparib 300 mg BID + lucitanib 4 mg QD (n=6)	Cohort 2: Rucaparib 400 mg BID + lucitanib 4 mg QD (n=4)	Cohort 3: Rucaparib 400 mg BID + lucitanib 6 mg QD (n=3)	Cohort 4: Rucaparib 600 mg BID + lucitanib 6 mg QD (n=6)	Overall (N=19)
Age, median (range), y	65.0 (51.0–76.0)	53.5 (36.0–68.0)	73.0 (65.0–73.0)	42.5 (39.0–53.0)	54.0 (36.0–76.0)
Female, n (%)	5 (83.3)	2 (50.0)	2 (66.7)	5 (83.3)	14 (73.7)
ECOG PS, n (%)					
0	1 (16.7)	0	1 (33.3)	0	2 (10.5)
1	5 (83.3)	4 (100)	2 (66.7)	6 (100)	17 (89.5)
Prior anticancer therapies, median (range)	3.5 (3.0–8.0)	4.0 (4.0–6.0)	4.0 (3.0–5.0)	3.0 (1.0–7.0)	4.0 (1.0–8.0)
Previous PARP inhibitor use, n (%)	1 (16.7)[a]	0	0	1 (16.7)[b]	2 (10.5)
Prior angiogenesis inhibitor use, n (%)	2 (33.3)	1 (25.0)	0	2 (33.3)	5 (26.3)
HRR gene mutation, n (%)[c]					
BRCA1	1 (16.7)	0	0	1 (16.7)	2 (10.5)
BRCA2	1 (16.7)	1 (25.0)	0	0	2 (10.5)
Other	3 (50.0)	0	2 (66.7)	0	5 (26.3)

Data cutoff date: April 16, 2021.
[a]Patient received olaparib as a fifth-line treatment for ovarian cancer. [b]Patient received olaparib as second-line maintenance for ovarian cancer. [c]Based on local genomics results from all patients and additional central Foundation Medicine, Inc., results available for patients in cohorts 1–3.
BID, twice daily; ECOG PS, Eastern Cooperative Oncology Group performance status; HRR, homologous recombination repair; PARP, poly(ADP-ribose) polymerase; QD, once daily.

Safety

- Median (range) time on treatment was 58.0 (20.0–395.0) days, with 3 patients ongoing as of the data cutoff date
- A DLT of grade 3 proteinuria was seen in cohort 1; no other DLTs have been reported
- Across all cohorts, the most common any-grade TEAEs were hypertension, nausea, and alanine aminotransferase (ALT)/aspartate aminotransferase (AST) increase (**Table 2**)
 - The most common grade ≥3 TEAE overall was anemia
- In addition to the DLT described above, 1 patient in cohort 1 had a TEAE (grade 3 hypertension) leading to treatment discontinuation

Table 2. TEAEs Occurring in ≥20% of Patients Overall

	Cohort 1: Rucaparib 300 mg BID + lucitanib 4 mg QD (n=6)		Cohort 2: Rucaparib 400 mg BID + lucitanib 4 mg QD (n=4)		Cohort 3: Rucaparib 400 mg BID + lucitanib 6 mg QD (n=3)		Cohort 4: Rucaparib 600 mg BID + lucitanib 6 mg QD (n=6)		Overall (N=19)	
n (%)	Any Grade	Grade ≥3	Any Grade	Grade ≥3	Any Grade	Grade ≥3	Any Grade	Grade ≥3	Any Grade	Grade ≥3
Patients with ≥1 TEAE	6 (100)	6 (100)	4 (100)	1 (25.0)	3 (100)	0	6 (100)	4 (66.7)	19 (100)	11 (57.9)
Hypertension	3 (50.0)	1 (16.7)	2 (50.0)	0	2 (66.7)	0	3 (50.0)	2 (33.3)	10 (52.6)	3 (15.8)
Nausea	3 (50.0)	0	2 (50.0)	1 (25.0)	2 (66.7)	0	2 (33.3)	0	9 (47.4)	1 (5.3)
ALT/AST increased	3 (50.0)	2 (33.3)	2 (50.0)	1 (25.0)	1 (33.3)	0	1 (16.7)	0	7 (36.8)	3 (15.8)
Anemia	2 (33.3)	2 (33.3)	2 (50.0)	1 (25.0)	0	0	2 (33.3)	1 (16.7)	6 (31.6)	4 (21.1)
Creatinine increased	2 (33.3)	0	1 (25.0)	0	0	0	3 (50.0)	0	6 (31.6)	0
Constipation	1 (16.7)	0	1 (25.0)	1 (25.0)	2 (66.7)	0	2 (33.3)	0	6 (31.6)	1 (5.3)
Asthenia/fatigue	2 (33.3)	1 (16.7)	1 (25.0)	0	1 (33.3)	0	1 (16.7)	0	5 (26.3)	1 (5.3)
Headache	2 (33.3)	0	0	0	1 (33.3)	0	2 (33.3)	0	5 (26.3)	0
Neutropenia[a]	1 (16.7)	1 (16.7)	2 (50.0)	0	1 (33.3)	0	1 (16.7)	0	5 (26.3)	1 (5.3)
Proteinuria	2 (33.3)	1 (16.7)	0	0	1 (33.3)	0	2 (33.3)	0	5 (26.3)	1 (5.3)
Thrombocytopenia[b]	1 (16.7)	1 (16.7)	1 (25.0)	0	1 (33.3)	0	2 (33.3)	0	5 (26.3)	1 (5.3)
ALP increased	1 (16.7)	1 (16.7)	0	0	1 (33.3)	0	2 (33.3)	1 (16.7)	4 (21.1)	2 (10.5)
Decreased appetite	2 (33.3)	0	0	0	0	0	2 (33.3)	0	4 (21.1)	0
Diarrhea	2 (33.3)	0	0	0	1 (33.3)	0	1 (16.7)	0	4 (21.1)	0
Hypomagnesemia	2 (33.3)	0	1 (25.0)	0	0	0	1 (16.7)	0	4 (21.1)	0
Hypothyroidism[c]	1 (16.7)	0	1 (25.0)	0	1 (33.3)	0	1 (16.7)	0	4 (21.1)	0
Insomnia	2 (33.3)	0	1 (25.0)	0	0	0	1 (16.7)	0	4 (21.1)	0

Data cutoff date: April 16, 2021.
[a]Includes neutropenia, neutrophil count decrease, and white blood cell count decrease. [b]Includes platelet count decrease and thrombocytopenia. [c]Includes hypothyroidism and blood thyroid-stimulating hormone increase.
ALP, alkaline phosphatase; ALT, alanine aminotransferase; AST, aspartate aminotransferase; BID, twice daily; QD, once daily; TEAE, treatment-emergent adverse event.

Pharmacokinetics

- Initial PK data were available for 13 patients in cohorts 1–3 at time of data cutoff (**Figure 1**)
 - Relatively large interpatient PK variability for rucaparib and lucitanib was observed; the phase 1/2 LIO-1 study (NCT04042116) is evaluating an individualized, safety-based dose-titration strategy with lucitanib, with the intended purpose of optimizing exposure and potential benefit of lucitanib-based combination treatment (see abstract 5538)
 - Lucitanib and rucaparib PK profiles in SEASTAR Arm A are consistent with those of previous monotherapy studies,[5,10] suggesting the lack of drug-drug interactions between these 2 agents

Figure 1. Plasma Concentration Time Profiles of (A) Rucaparib and (B) Lucitanib

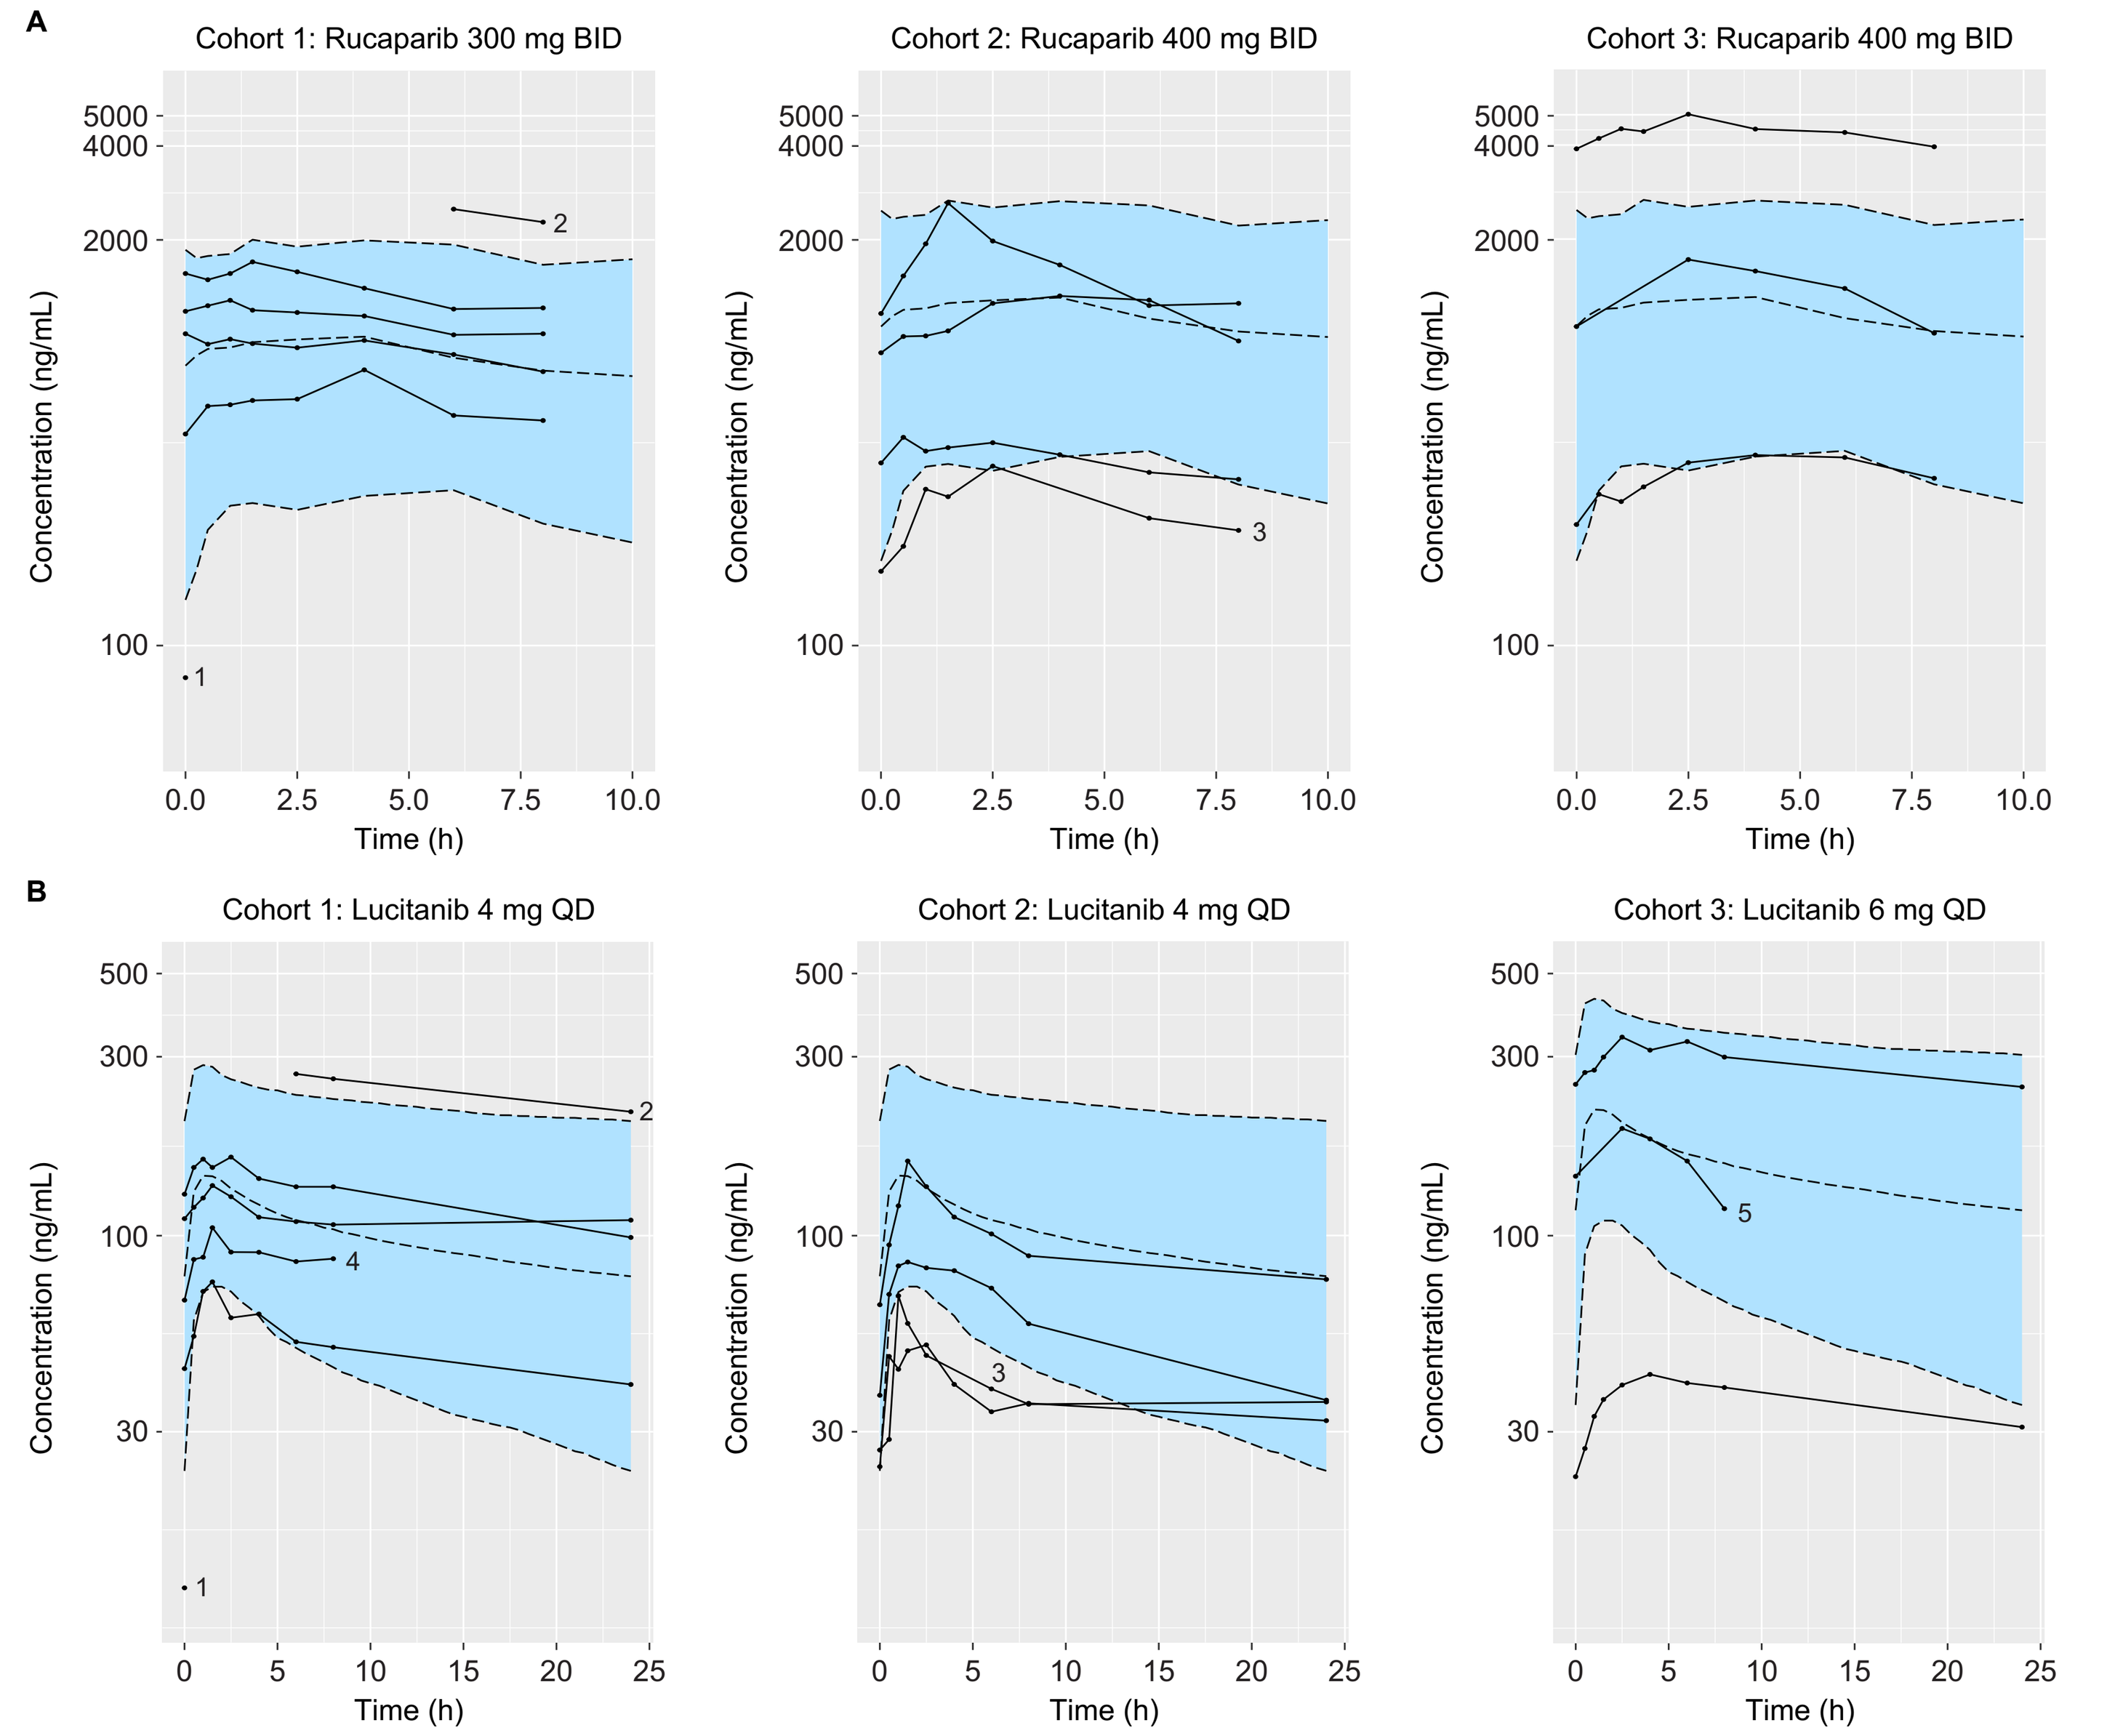
A
Cohort 1: Rucaparib 300 mg BID
Cohort 2: Rucaparib 400 mg BID
Cohort 3: Rucaparib 400 mg BID
Concentration (ng/mL)
Time (h)
B
Cohort 1: Lucitanib 4 mg QD
Cohort 2: Lucitanib 4 mg QD
Cohort 3: Lucitanib 6 mg QD
Concentration (ng/mL)
Time (h)

Each solid line represents an individual patient. In the blue shaded area, dashed lines represent median and 90% prediction interval of simulated PK data. (A) Rucaparib PK data were simulated using PK data from Study 10 (NCT01482715) dose-normalized to 300 or 400 mg BID.[10] (B) Lucitanib PK data were simulated for 500 subjects receiving 4 or 6 mg QD using a population PK model developed on previous monotherapy data.[5] Patient 1 experienced a DLT; only the predose PK sample was collected. For patient 2, some PK samples were not collected. Patient 3 took half doses of drugs until cycle 1 day 15. For patients 4 and 5, 24-h postdose lucitanib PK samples were not collected.
BID, twice daily; DLT, dose-limiting toxicity; PK, pharmacokinetics; QD, once daily.

Efficacy

- Among patients with measurable disease, 1 patient in cohort 1 with *PALB2*-mutated advanced endometrial cancer had a confirmed partial response per RECIST, lasting 30.3 weeks (**Table 3**, **Figure 2**)
 - Six patients had a best overall response of stable disease, including 1 patient in cohort 1 with an unconfirmed PR, and 1 patient each in cohorts 1, 3, and 4, with stable disease ≥16 weeks
- One patient in cohort 2 with *BRCA2*-mutated castration-resistant prostate cancer continued to receive treatment despite initial progressing bone metastases (**Figure 2**), resulting in a prostate-specific antigen response (≥50% change) lasting 32+ weeks and a best change in sum of target lesions of −52.2%

Table 3. ORR and Disease Control Rate

	Cohort 1: Rucaparib 300 mg BID + lucitanib 4 mg QD (n=5)	Cohort 2: Rucaparib 400 mg BID + lucitanib 4 mg QD (n=4)	Cohort 3: Rucaparib 400 mg BID + lucitanib 6 mg QD (n=3)	Cohort 4: Rucaparib 600 mg BID + lucitanib 6 mg QD (n=5)	Overall (N=17)
Confirmed RECIST ORR, n (%) [95% CI]	**1 (20.0) [0.5–71.6]**	**0**	**0**	**0**	**1 (5.9) [0.1–28.7]**
Complete response	0	0	0	0	0
Partial response	1 (20.0)	0	0	0	1 (5.9)
Stable disease	3 (60.0)[a]	0	2 (66.7)	1 (20.0)	6 (35.3)
Progressive disease	1 (20.0)	4 (100)	1 (33.3)	1 (20.0)	7 (41.2)
Not evaluable	0	0	0	3 (60.0)[b]	3 (17.6)
Disease control rate, n (%) [95% CI][c]	**2 (40.0) [5.3–85.3]**	**0 [0–60.2]**	**1 (33.3) [0.8–90.6]**	**1 (20.0) [0.5–71.6]**	**4 (23.5) [6.8–49.9]**

Data cutoff date: April 16, 2021.
[a]Patient had an unconfirmed partial response. [b]Includes 2 patients ongoing on study for whom a follow-up scan was not yet available. [c]Defined as a complete response, partial response, or stable disease ≥16 weeks.
BID, twice daily; ORR, objective response rate; QD, once daily; RECIST, Response Evaluation Criteria In Solid Tumors, version 1.1.

Figure 2. Duration of Treatment and Best Overall Response to Rucaparib + Lucitanib to Date

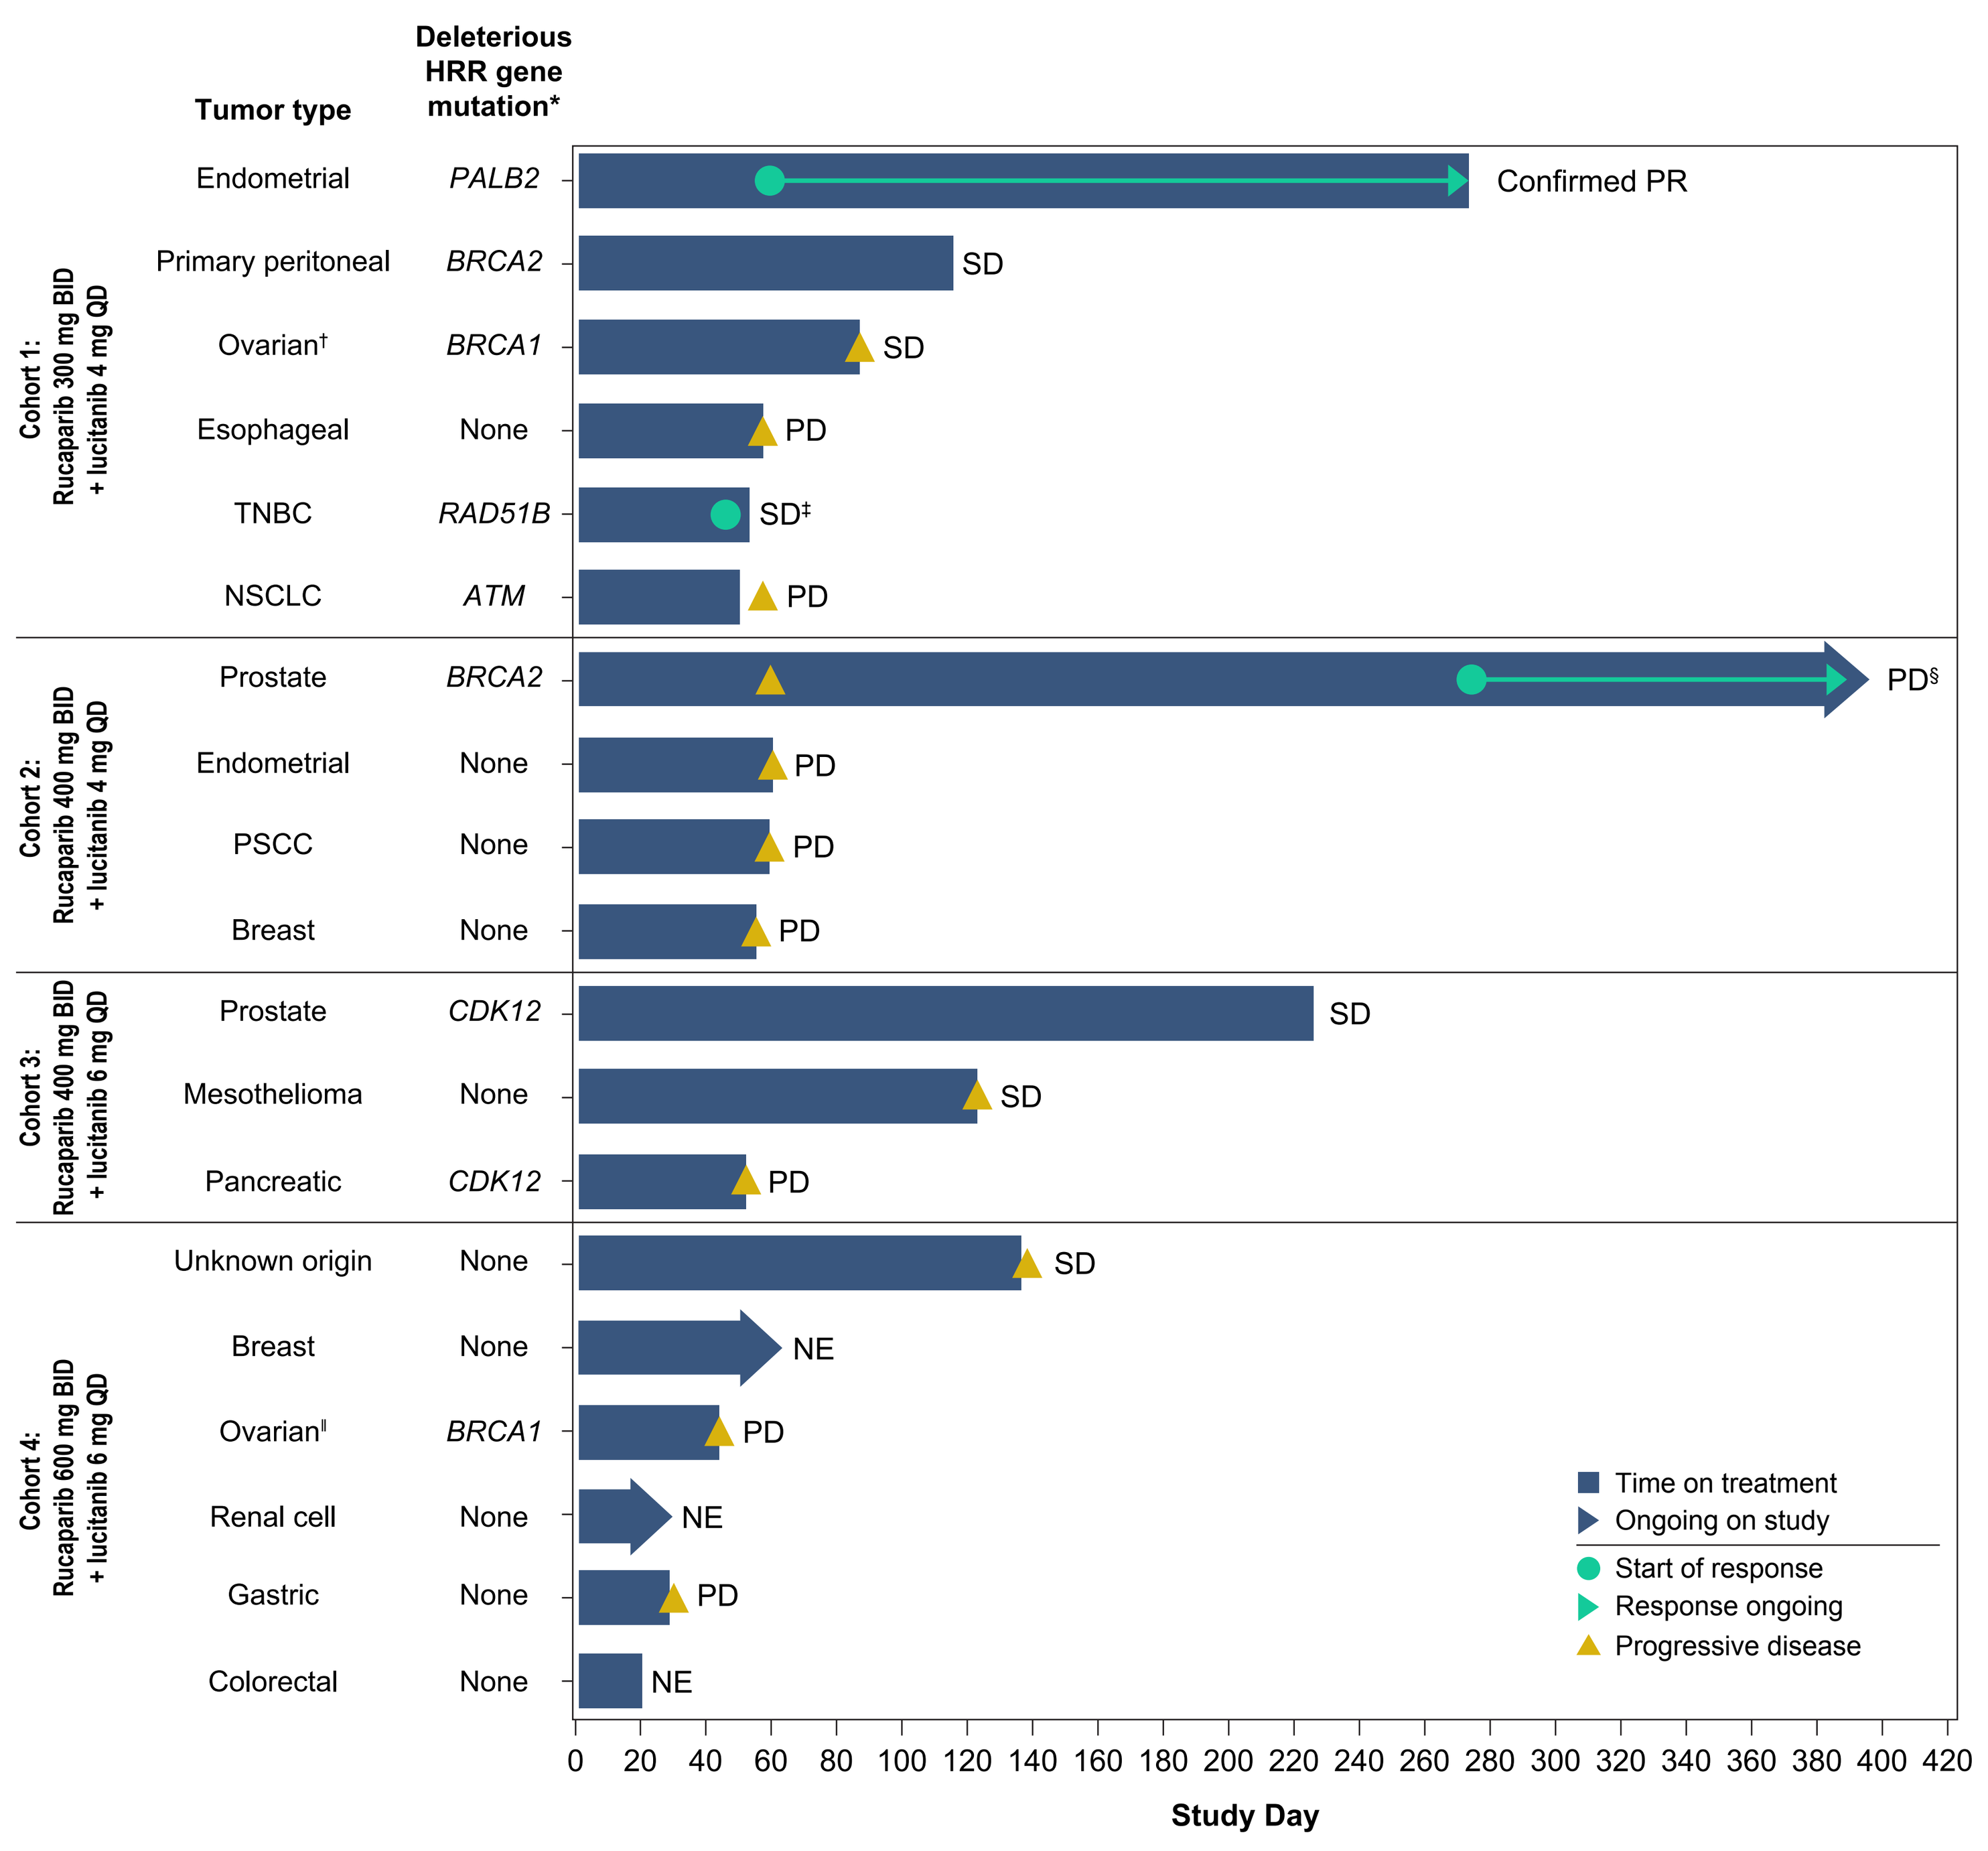
Tumor type
Deleterious HRR gene mutation*
Cohort 1: Rucaparib 300 mg BID + lucitanib 4 mg QD
Endometrial PALB2 Confirmed PR
Primary peritoneal BRCA2 SD
Ovarian† BRCA1 SD
Esophageal None PD
TNBC RAD51B SD‡
NSCLC ATM PD
Cohort 2: Rucaparib 400 mg BID + lucitanib 4 mg QD
Prostate BRCA2 PD§
Endometrial None PD
PSCC None PD
Breast None PD
Cohort 3: Rucaparib 400 mg BID + lucitanib 6 mg QD
Prostate CDK12 SD
Mesothelioma None SD
Pancreatic CDK12 PD
Cohort 4: Rucaparib 600 mg BID + lucitanib 6 mg QD
Unknown origin None SD
Breast None NE
Ovarian‖ BRCA1 PD
Renal cell None NE
Gastric None PD
Colorectal None NE
Time on treatment
Ongoing on study
Start of response
Response ongoing
Progressive disease
0 20 40 60 80 100 120 140 160 180 200 220 240 260 280 300 320 340 360 380 400 420
Study Day

Data cutoff date: April 16, 2021. *Based on local genomics results from all patients and additional central Foundation Medicine, Inc., results available for patients in cohorts 1–3. †Patient received olaparib as a fifth-line treatment for ovarian cancer. ‡Patient has an unconfirmed PR. §Patient continued to receive treatment despite initial PD and subsequently had a reduction in sum of target lesions over the –30% threshold for PR at the week 40 scan. ‖Patient received olaparib as second-line maintenance for ovarian cancer.
BID, twice daily; HRR, homologous recombination repair; NE, not evaluable; NSCLC, non–small cell lung cancer; PD, progressive disease; PR, partial response; PSCC, papillary squamous cell carcinoma; QD, once daily; SD, stable disease; TNBC, triple-negative breast cancer.

REFERENCES

1. Wahlberg et al. *Nat Biotechnol*. 2012;30:283-8.
2. Thomas et al. *Mol Cancer Ther.* 2007;6:945-56.
3. Robillard et al. *Cancer Res*. 2017;77(suppl 13):abst 2475.
4. Bello et al. *Cancer Res*. 2011;71:1396-405.
5. Data on file. Clovis Oncology, Inc. 2021.
6. Bindra et al. *Cancer Res*. 2005;65:11597-604.
7. Hegan et al. *Proc Natl Acad Sci U S A*. 2010;107:2201-6.
8. Chan et al. *Cancer Res*. 2010;70:8045-54.
9. Lim et al. *Neoplasia*. 2014;16:343-53.e1-2.
10. Shapiro et al. *Clin Pharmacol Drug Dev*. 2019;8:107-18.

ACKNOWLEDGMENTS

This study was funded by Clovis Oncology, Inc. Medical writing and editorial support funded by Clovis Oncology, Inc., were provided by Nathan Yardley and Stephen Bublitz of Ashfield MedComms, an Ashfield Health company.

ASCO Annual Meeting | June 4–8, 2021 | Virtual Meeting